IRAK4 inhibitors display synergistic activity when combined with BTK or PI3K inhibitors in B-cell lymphomas

Eric G. Vajda[1], Robert Niecestro[2], Lin Zhi[1], and Keith B. Marschke[1]
[1]Ligand Pharmaceuticals, La Jolla, CA [2]TG Therapeutics, New York, NY

Abstract # 785

Background

- Interleukin-1 Receptor Associated Kinase 4 (IRAK4) is a serine/threonine protein kinase that plays an essential role in interleukin-1 receptor and toll-like receptor signaling pathways
- IRAK4 forms a complex with Myeloid Differentiation Primary Response Gene 88 (MYD88) that facilitates downstream signaling
- Recent studies have identified oncogenically active mutations in the MYD88 gene in human B-cell lymphomas
- IRAK4 inhibition has been proposed as a potential treatment for B-cell lymphomas, particularly those containing mutations in MYD88
- We have investigated the effects of novel IRAK4 inhibitors on B-cell proliferation and apoptosis both as single agents and in combination with other kinase inhibitors

Methods

- **Kinase inhibition assays**: Kinase inhibition assays were performed by Reaction Biology Corporation utilizing a ^{33}P-ATP radioisotope filtration binding technique.
- **Interleukin-1 inhibition assay**: Inhibition of the IL-1 pathway was performed in the A549 cell line. Cells were seeded into 96 well plates and compounds were added in dose response with DMSO as a control. Cells were stimulated overnight with 10 ng/mL recombinant IL-1β. IL-6 concentration of the supernatant was measured using an AlphaLISA assay.
- **IRAK1 degradation**: Inhibition of IRAK1 degradation was examined in A549 cells. Cells were pre-treated for 15 minutes with compounds followed by 10 minute treatment with recombinant IL-1β. Whole cell lysates were analyzed by Western blot for IRAK1 and β-actin levels.
- **Lymphoma cell lines**: Four B-cell lymphoma cell lines were selected for viability, apoptosis, and synergism studies. Cells were cultured in growth media as described by the vendors. OCI-LY19 cells (B-cell non-hodgkin lymphoma/DLCL) and OCI-LY3 cells (B-cell non-hodgkin lymphoma/DLBCL) were obtained from DSMZ. U266B1 (lymphoblastic plasmyacytoma) was obtained from ATCC and MWCL1 cells (Waldenström macroglobulinemia) from the Mayo Foundation for Medical Education and Research. Cells contained either a WT MYD88 adaptor protein (OCI-LY19, U266) or a L265P mutation in MYD88 (OCI-LY3, MWCL1).
- **Cell viability assays**: Cell viability was examined using the ATPLite luminescent assay (PerkinElmer). Cells were incubated in 96 well plates for 72 hours in the presence of compounds or DMSO.
- **Apoptosis assays**: Cells were incubated at 37°C with an IRAK4 inhibitor for either 4 or 24 hours at a concentration equal to the IC_{50} value determined in the viability assays. Cells were stained with either Annexin V/propidium iodide or Caspase 3&7/Sytox dead cell stain (Life Technologies). The dual stains were examined by flow cytometry to determine the number of viable, dead, or apoptotic cells.
- **Synergism studies**: Cells were treated with IRAK4 inhibitors and either BTK or PI3K inhibitors in combination. Compounds were added in a constant-ratio dose response and cell viability was determined by ATPLite assay.

Results

Compounds

Kinase Inhibition

- LG0224912 is a potent small molecule, IRAK4 inhibitor with an IC_{50} = 0.7 nM in kinase inhibition assays
- LG0224912 has selective activity against a broad range of kinases[1] but does inhibit IRAK1, FLT3, CLK2, and DYRK1B (<10 fold selective)
- LG0250276 is a potent small molecule, IRAK4 inhibitor with an IC_{50} = 2.7 nM in kinase inhibition assays and improved selectivity vs FLT3, CLK3, and DYRK1B

Inhibition of Interleukin-1 Signaling

- IL-1 stimulates IL-6 secretion in A549 cells through an IRAK4 mediated pathway. Inhibition of IL-6 secretion was used to assess compound effects in a whole cell assay
- Compounds potently inhibited IL-6 production in A549 cells
 LG0224912 IC_{50}= 90 nM LG0250276 IC_{50}= 79 nM

Inhibition of IRAK1 degradation

- IL-1 signaling leads to IRAK4 phosphorylation and suqsequent IRAK1 ubiquitination and proteosomal degradation.
- LG0224912 inhibits IRAK1 degradation in IL-1 stimulated A549 cells as determined by Western blot

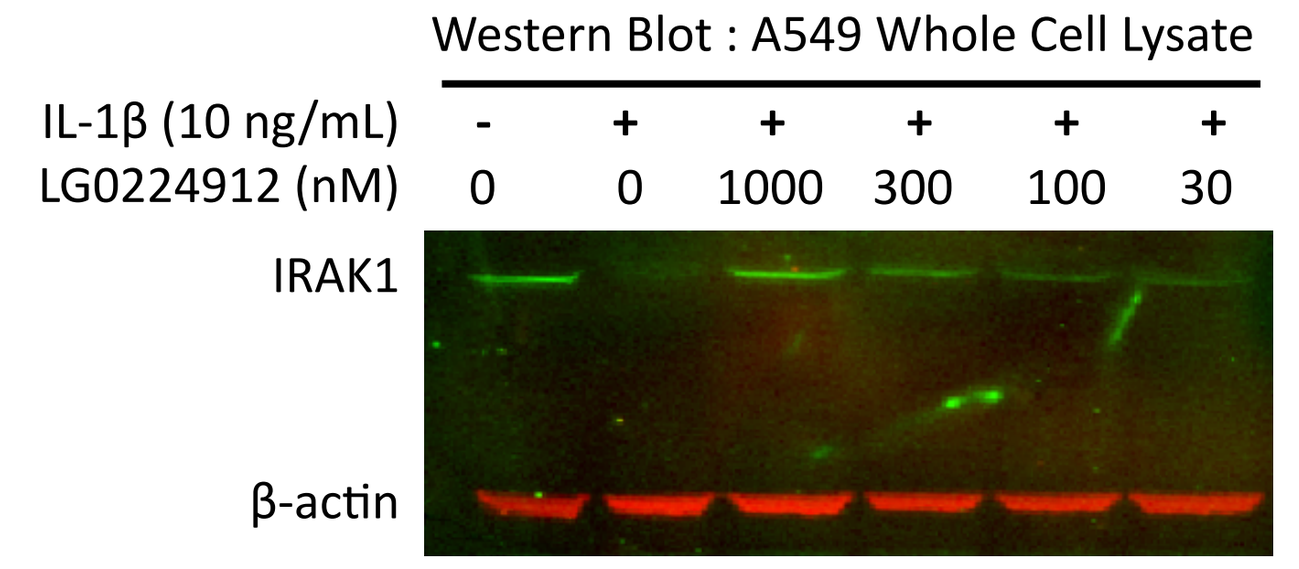
Western Blot : A549 Whole Cell Lysate
IL-1β (10 ng/mL) - + + + + +
LG0224912 (nM) 0 0 1000 300 100 30
IRAK1
β-actin

Anti-proliferative Effects

- Cell viability was assessed in cell lines with WT MYD88 (OCI-LY19 & U266) and cell lines with L265P mutated MYD88 (MWCL1 & OCI-LY3)
- After 72 hours, cell viability was reduced in both WT and mutated cell lines
- Representative graphs from two cell lines are shown and the data from replicate experiments are presented in the table

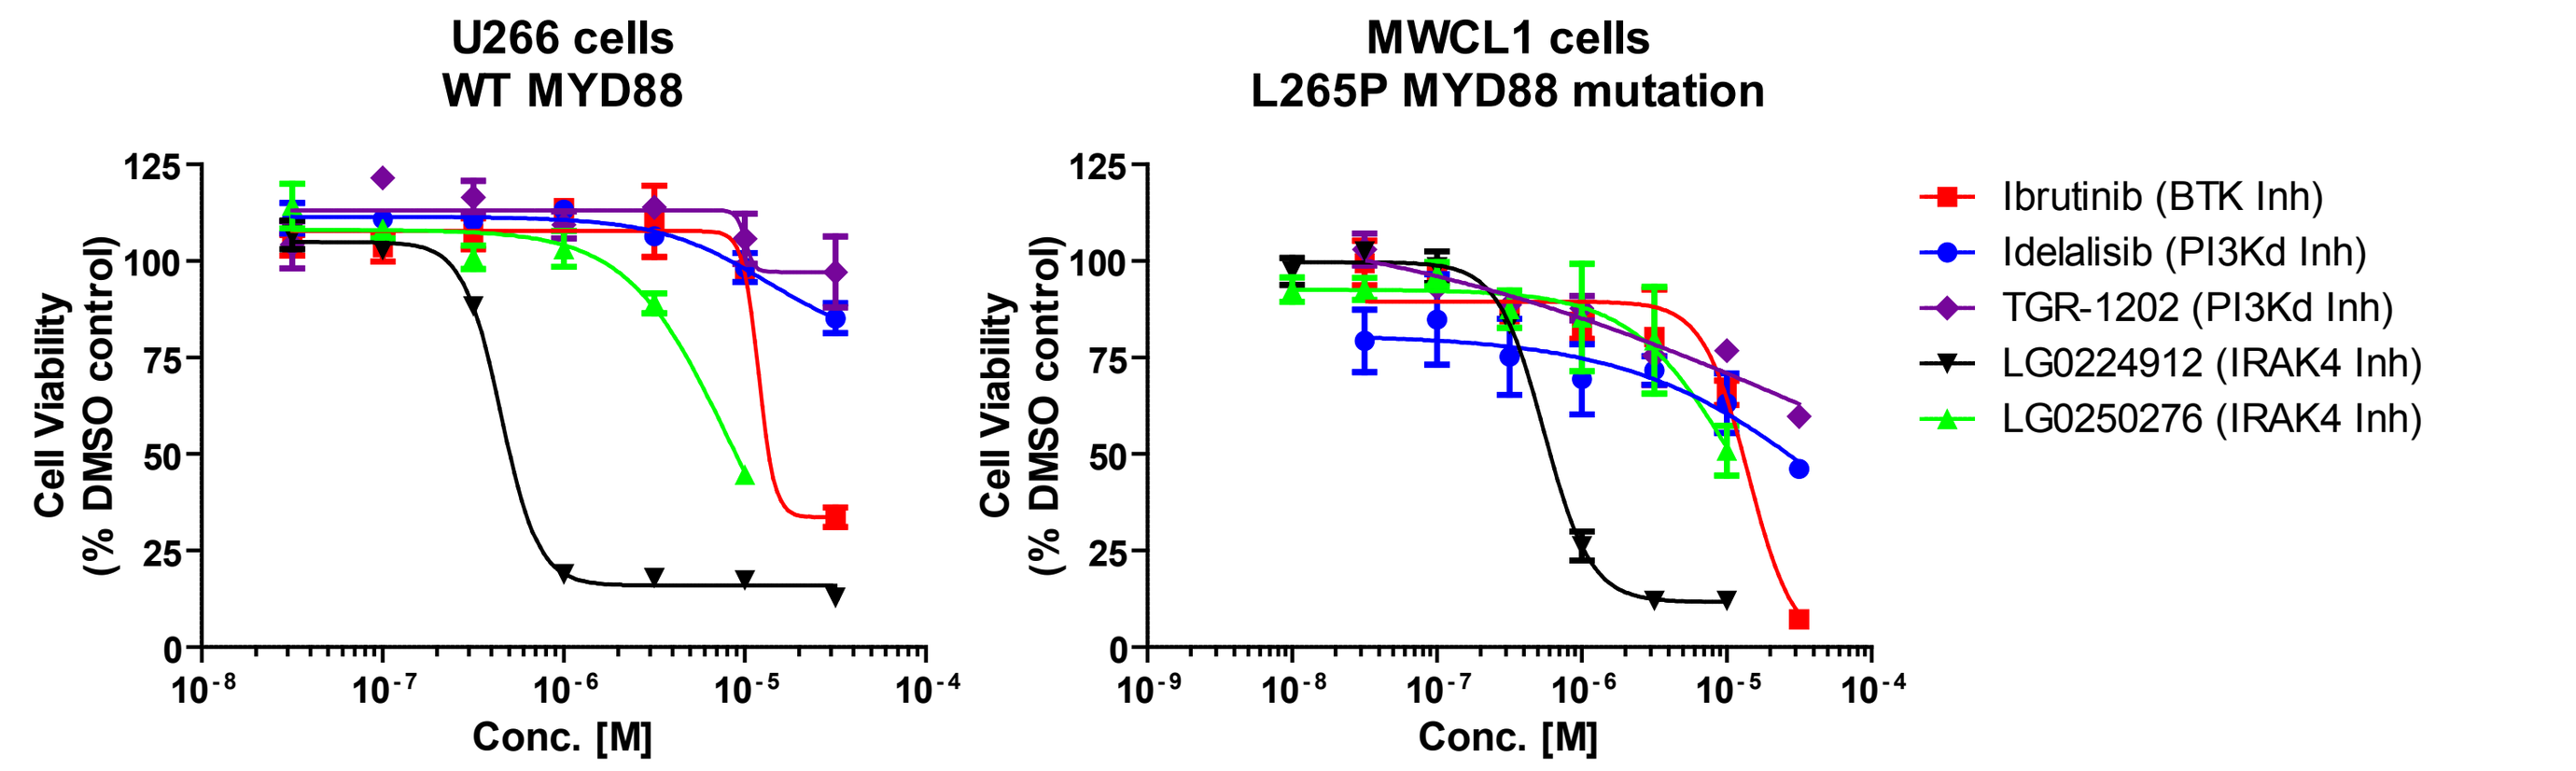
U266 cells
WT MYD88
MWCL1 cells
L265P MYD88 mutation
Cell Viability
(% DMSO control)
Conc. [M]
Ibrutinib (BTK Inh)
Idelalisib (PI3Kd Inh)
TGR-1202 (PI3Kd Inh)
LG0224912 (IRAK4 Inh)
LG0250276 (IRAK4 Inh)

		OCI-LY3					OCI-LY19					MWCL1					U266				
	Primary	Potency (nM)		Efficacy (%)			Potency (nM)		Efficacy (%)			Potency (nM)		Efficacy (%)			Potency (nM)		Efficacy (%)		
Compound	Target	Mean	SD	Mean	SD	n	Mean	SD	Mean	SD	n	Mean	SD	Mean	SD	n	Mean	SD	Mean	SD	n
Ibrutinib	BTK	14347	3632	94	2	3	11517	670	99	1	3	13963	1312	93	1	3	20720	8894	58	8	3
Idelalisib	PI3K	>30000	--	48	21	3	>30000	--	42	1	3	>30000	--	49	6	3	>30000	--	12	3	3
TGR-1202	PI3K	7631	7833	79	7	2	>30000	--	40	3	2	>30000	--	40	1	2	>30000	--	6	5	2
LG0224912	IRAK4	567	252	100	0	3	397	21	100	0	4	475	88	87	2	3	378	62	86	1	3
LG0250276	IRAK4	>10000	--	38	27	3	8025	6054	54	16	3	>10000	--	44	15	3	>10000	--	32	20	3

Apoptosis

- To investigate the mechanisms responsible for the anti-proliferative effects of IRAK4 inhibitors, apoptosis was examined using flow cytometry
- Apoptosis was assessed by two complimentary techniques: propidium iodide/AnnexinV dual staining or Sytox Dead Cell Stain/Caspase 3&7 dual staining
- Representative dot plots in the OCI-LY19 cell line demonstrate increased apoptosis at early time points and cell death at later timepoints after treatment with IRAK4 inhibitors (blue=viable cells, green=apoptotic cells, red=dead cells).
- Graphs represent means from 3 separate experiments
- Similar results were observed for the OCI-LY3 cell line (data not shown)

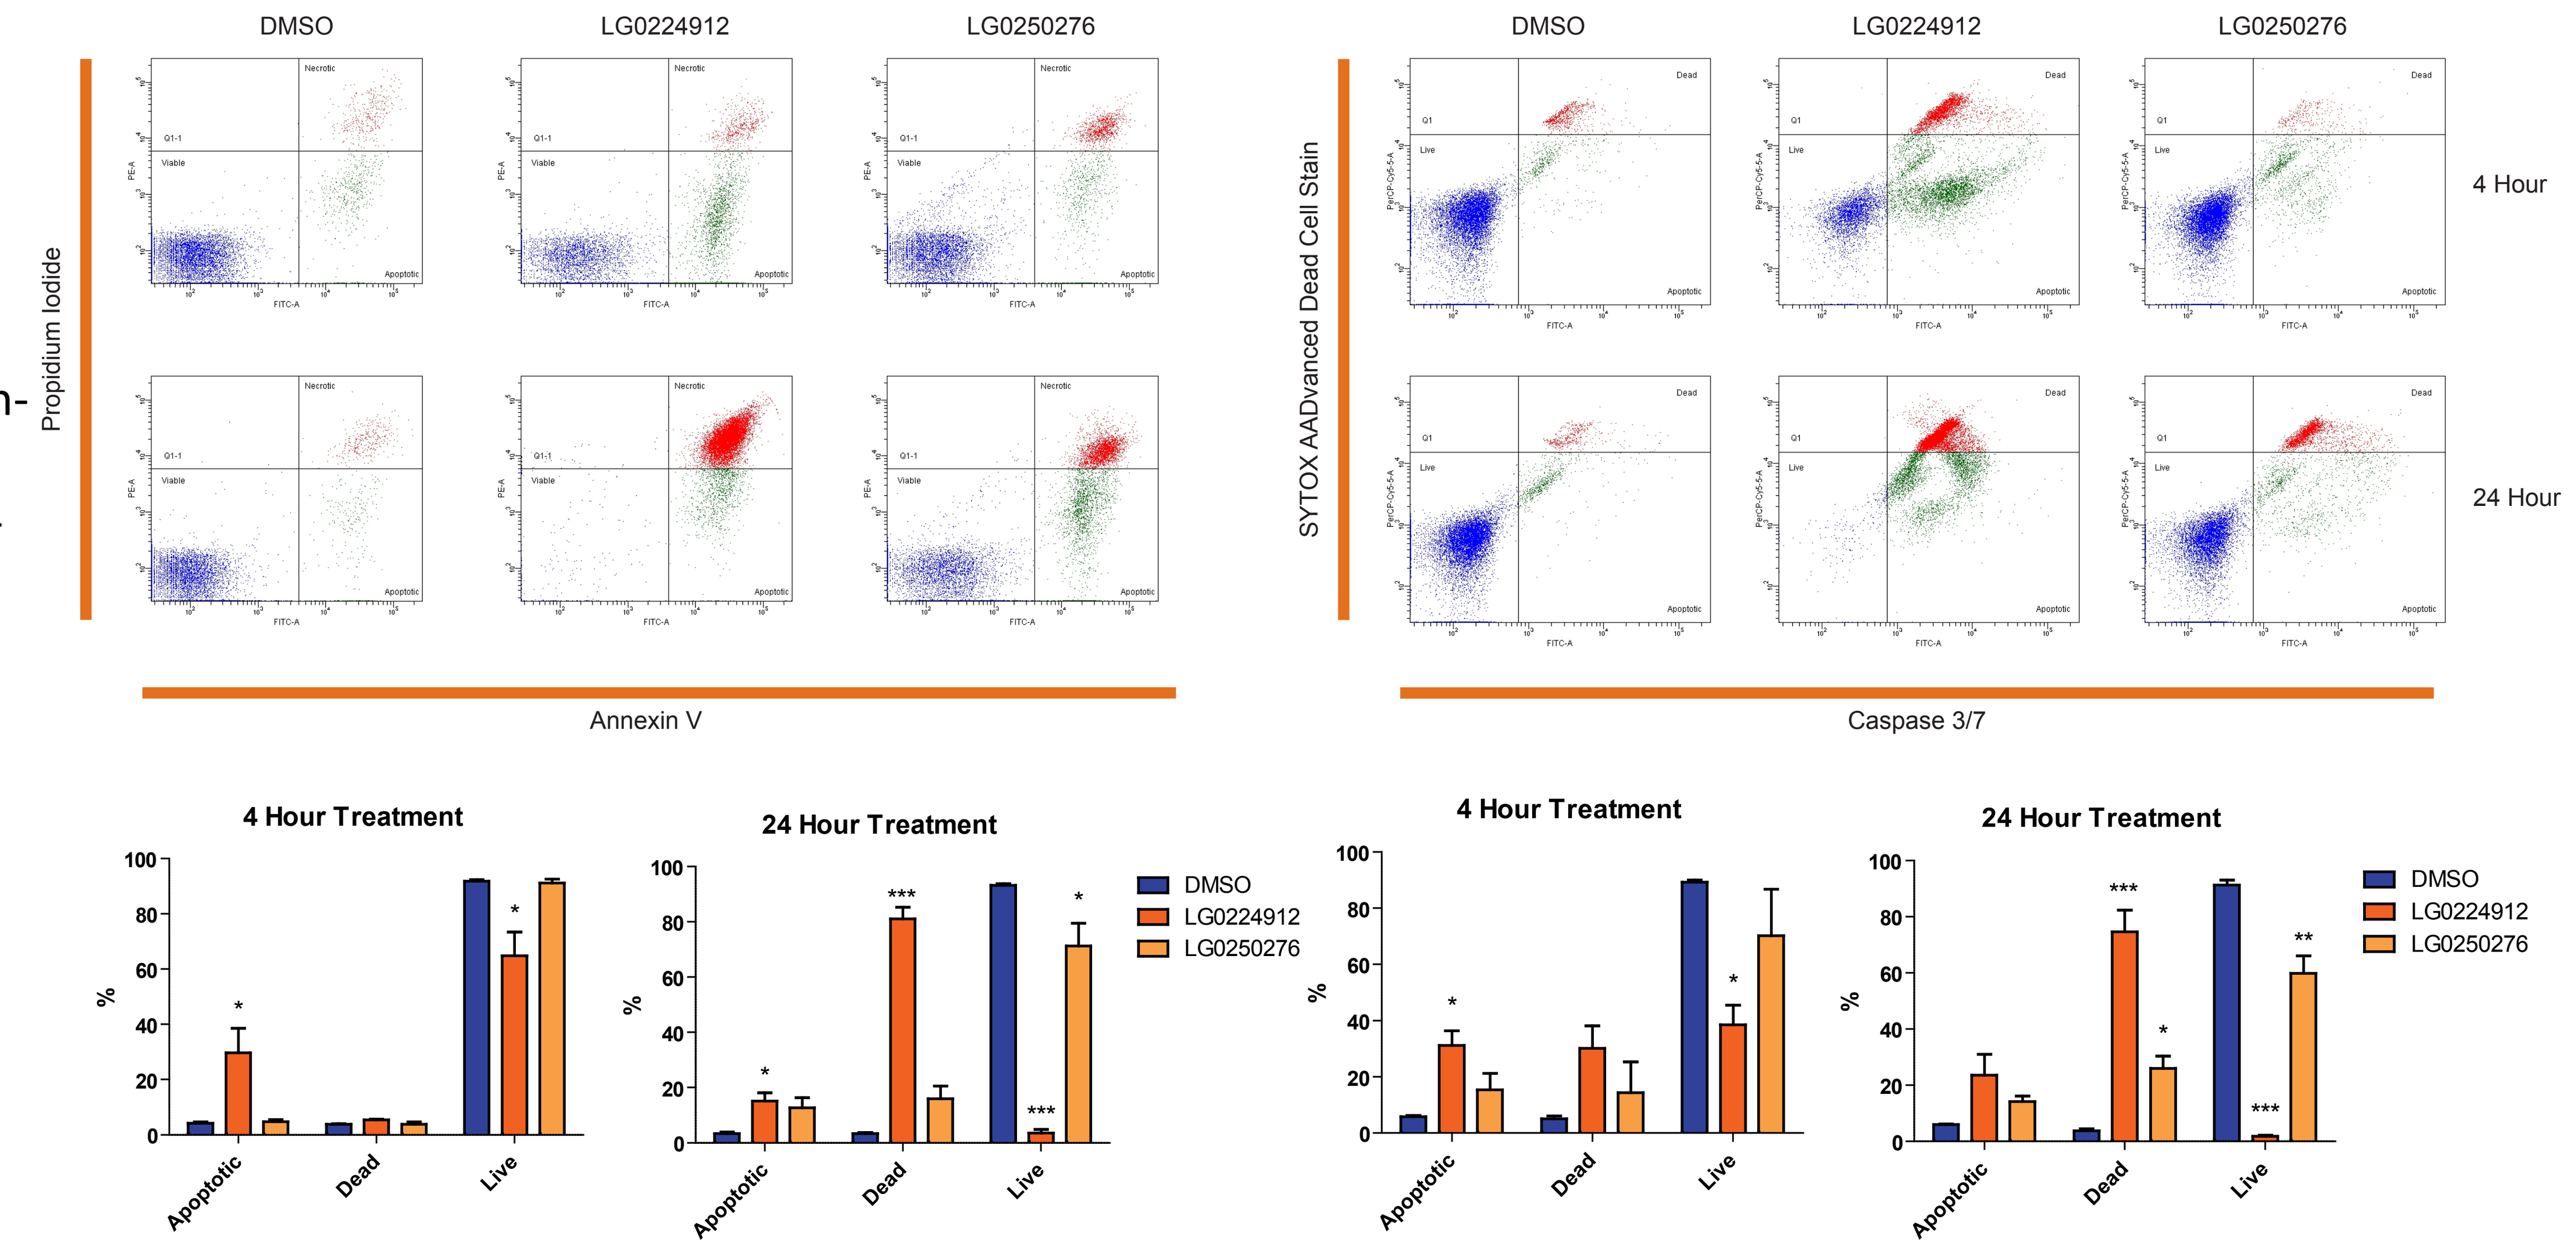
DMSO
LG0224912
LG0250276
Propidium Iodide
Annexin V
SYTOX AADvanced Dead Cell Stain
Caspase 3/7
4 Hour
24 Hour
4 Hour Treatment
24 Hour Treatment
%
Apoptotic
Dead
Live
DMSO
LG0224912
LG0250276

Synergistic Effects

- Cell viability was examined with IRAK4 inhibitors in combination with either ibrutinib (BTK inhibitor) or TGR-1202 (PI3Kδ inhibitor)
- Compounds were administered in dose response with a constant molar ratio between inhibitors and the cell viability was measured using the ATPLite assay
- Cell viability was normalized from 0 to 1 to yield the fractional affect (Fa)
- Combination index (CI) was calculated using CompuSyn software to define synergism or antagonism following the method of Chou-Talalay[2]
- Combination treatment with LG0250276 and either ibrutinib or TGR-1202 was synergistic at higher concentrations
- Combination treatment with LG0224912 did not demonstrate significant synergism and combinations were generally additive (data not shown)

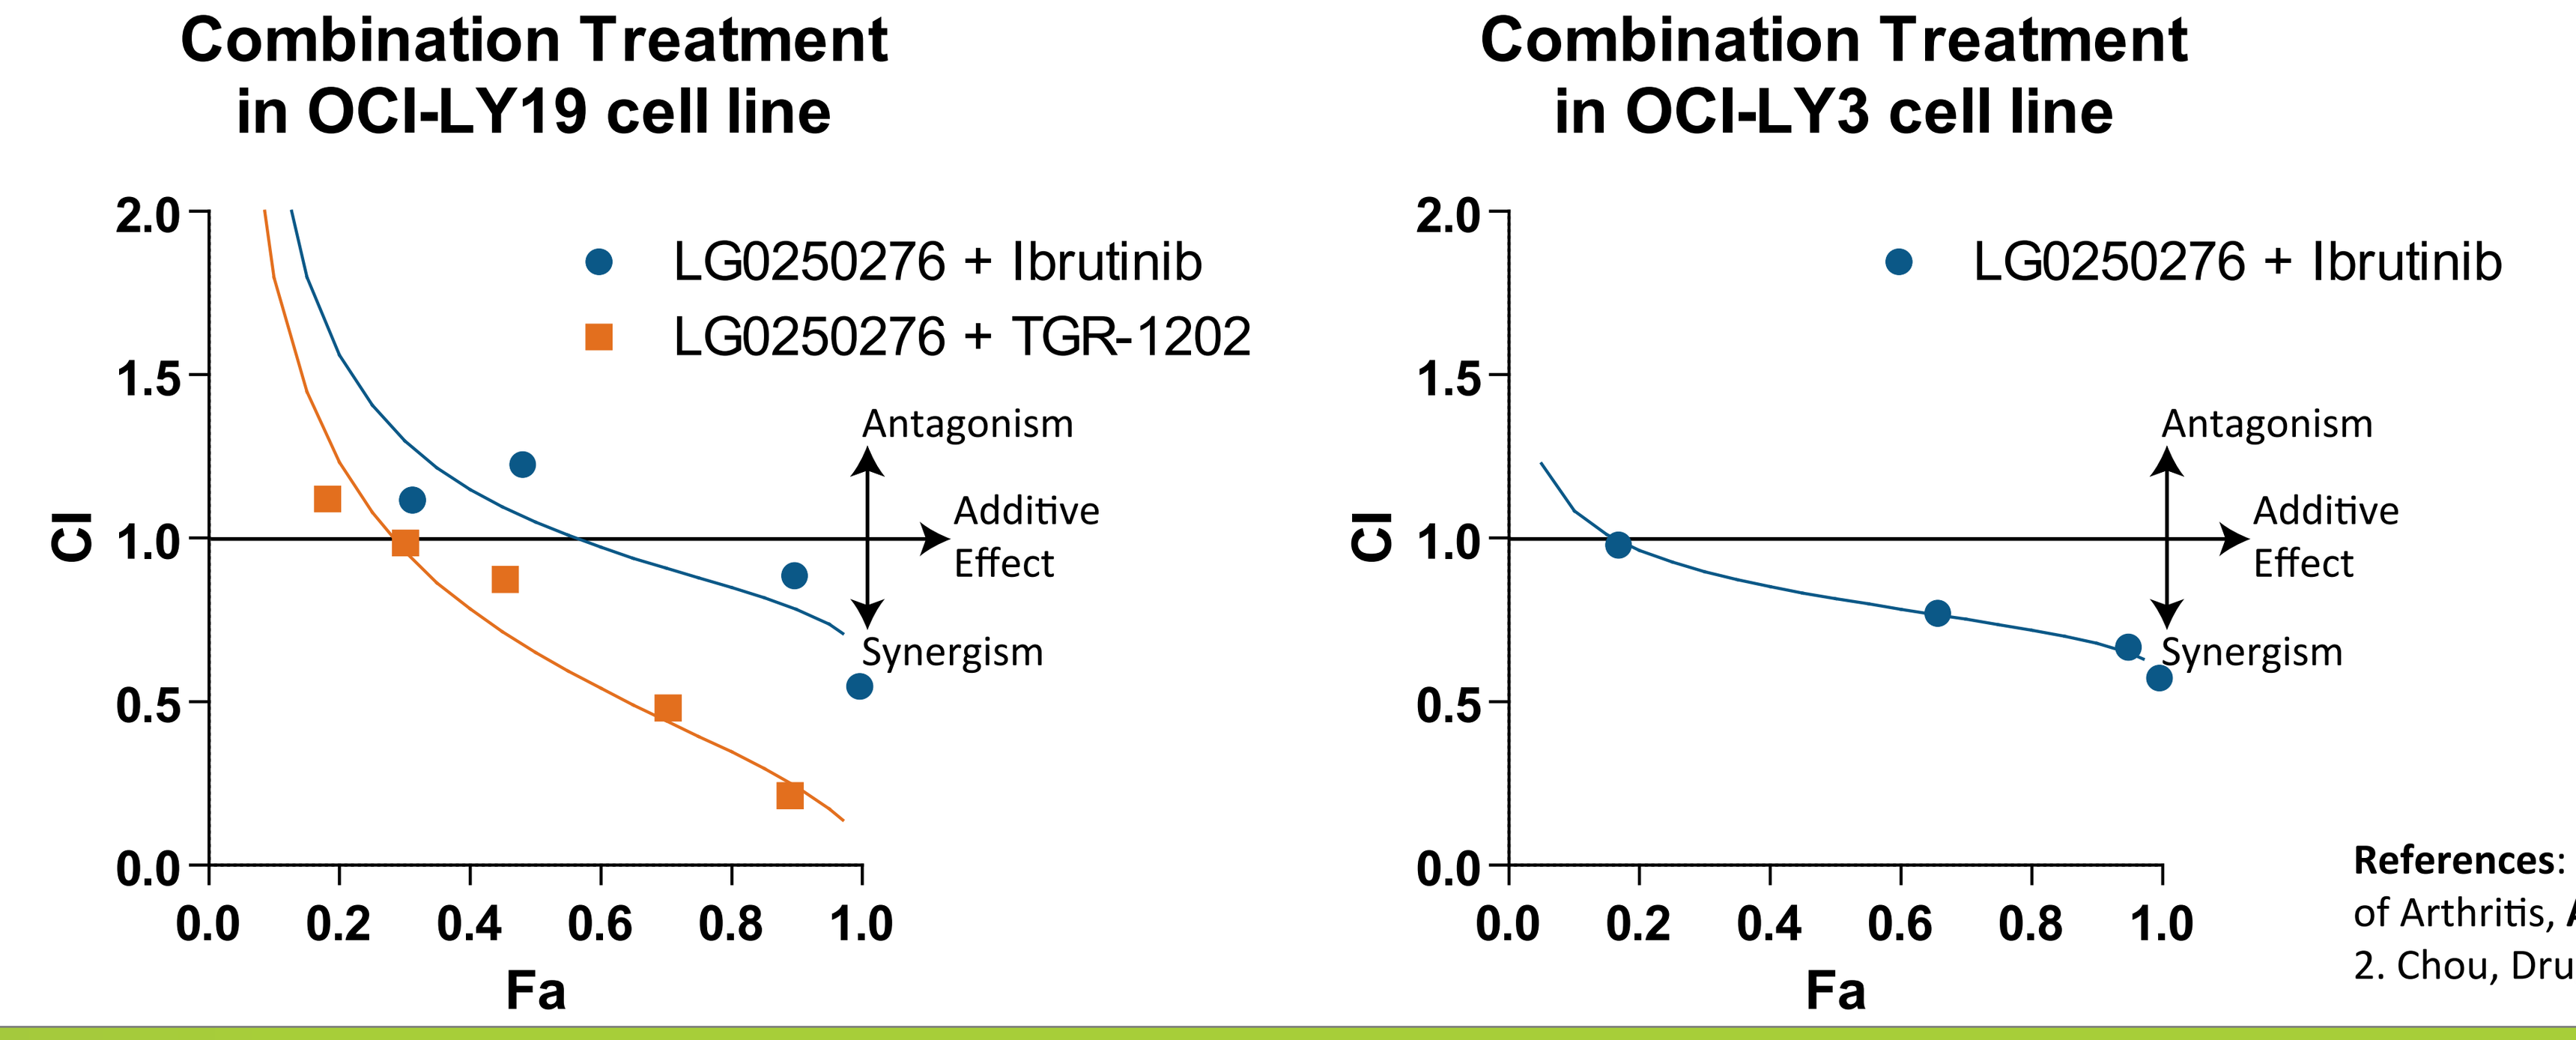
Combination Treatment
in OCI-LY19 cell line
LG0250276 + Ibrutinib
LG0250276 + TGR-1202
Antagonism
Additive
Effect
Synergism
CI
Fa
Combination Treatment
in OCI-LY3 cell line
LG0250276 + Ibrutinib

Conclusions

- Potent small molecule IRAK4 inhibitors have been identified that block IRAK4 and IL-1 mediated signaling in cell based models
- IRAK4 inhibitors reduce proliferation in B-cell lymphomas in the presence of WT or mutated MYD88 adaptor protein
- The anti-proliferative effects of IRAK4 inhibitors are mediated in part by increased apoptosis
- Combinations of IRAK4 inhibitors with ibrutinib (BTK inhibitor) or TGR-1202 (PI3Kδ inhibitor) have synergistic activity in B-cell lymphomas
- IND-enabling nonclinical toxicity studies are underway

References: 1. Vajda *et al.*, Novel Small Molecule Inhibitors of Interleukin-1 Receptor Associated Kinase-4 Are Effective in a Preclinical Model of Arthritis, **American College of Rheumatology Annual Meeting**, 2011.
2. Chou, Drug Combination Studies and Their Synergy Quantification Using the Chou-Talalay Method, **Cancer Res.** 2010;70:440-446.

Presented at the American Association for Cancer Research (AACR) Annual Meeting, April 18-April 22, 2015, Philadelphia, PA