Abstract # 4421

The PI3K Delta Inhibitor TGR-1202 and Proteasome Inhibitor Carfilzomib Are Highly Synergistic in Killing Human B- and T- Cell Lymphoma Cells

Changchun Deng[1], Mark Lipstein[1], Christine McIntosh[2], Richard Rodriguez[1], Peter Sportelli[3], Hari P. Miskin[3], Swaroop Vakkalanka[3], Srikant Viswanadha[4], Owen A. O'Connor[1]

COLUMBIA UNIVERSITY MEDICAL CENTER

[1]Center for Lymphoid Malignancies, Department of Medicine, Columbia University Medical Center, New York, NY; [2]College of Arts and Sciences, Cornell University, New York; [3]TG Therapeutics, Inc., New York, NY; [3]Rhizen Pharmaceuticals, La Chaux-de-Fonds, Switzerland; [4]Incozen Therapeutics, Hyderabad, India

BACKGROUND

Constitutively activated PI3K/AKT/mTOR pathway plays a key role in the proliferation and survival of cancer cells. Inhibitors of PI3K-δ such as TGR-1202 and GS-1101 (idelalisib) have shown promising activity in the treatment of B cell lymphoma. On the other hand, T cell lymphoma (TCL) cells are effectively inhibited only when both PI3K-δ and PI3K-γ are inhibited or knocked-down (Subramaniam et al., Cancer Cell 21, 2012). Carfilzomib is an irreversible proteasome inhibitor that potently inhibits the activation of NF-κB by preventing degradation of the NF-κB inhibitor IκB. We hypothesize that if both PI3K/AKT and proteasome are involved in the activation of the pro-survival pathways, then combining a PI3K-δ inhibitor and a proteasome inhibitor will synergistically inhibit the growth of lymphoma cells. Here we present data demonstrating that TGR-1202 and carfilzomib are markedly synergistic in models of B- and T- cell lymphoma. We also present preliminary results demonstrating that the synergism resulted from the enhanced stimulatory effect that these two drugs in combination have on the expression of p53-responsive genes, such as p21 and Bax, and on the cleavage of poly(ADP-ribose) polymerase.

RESULTS

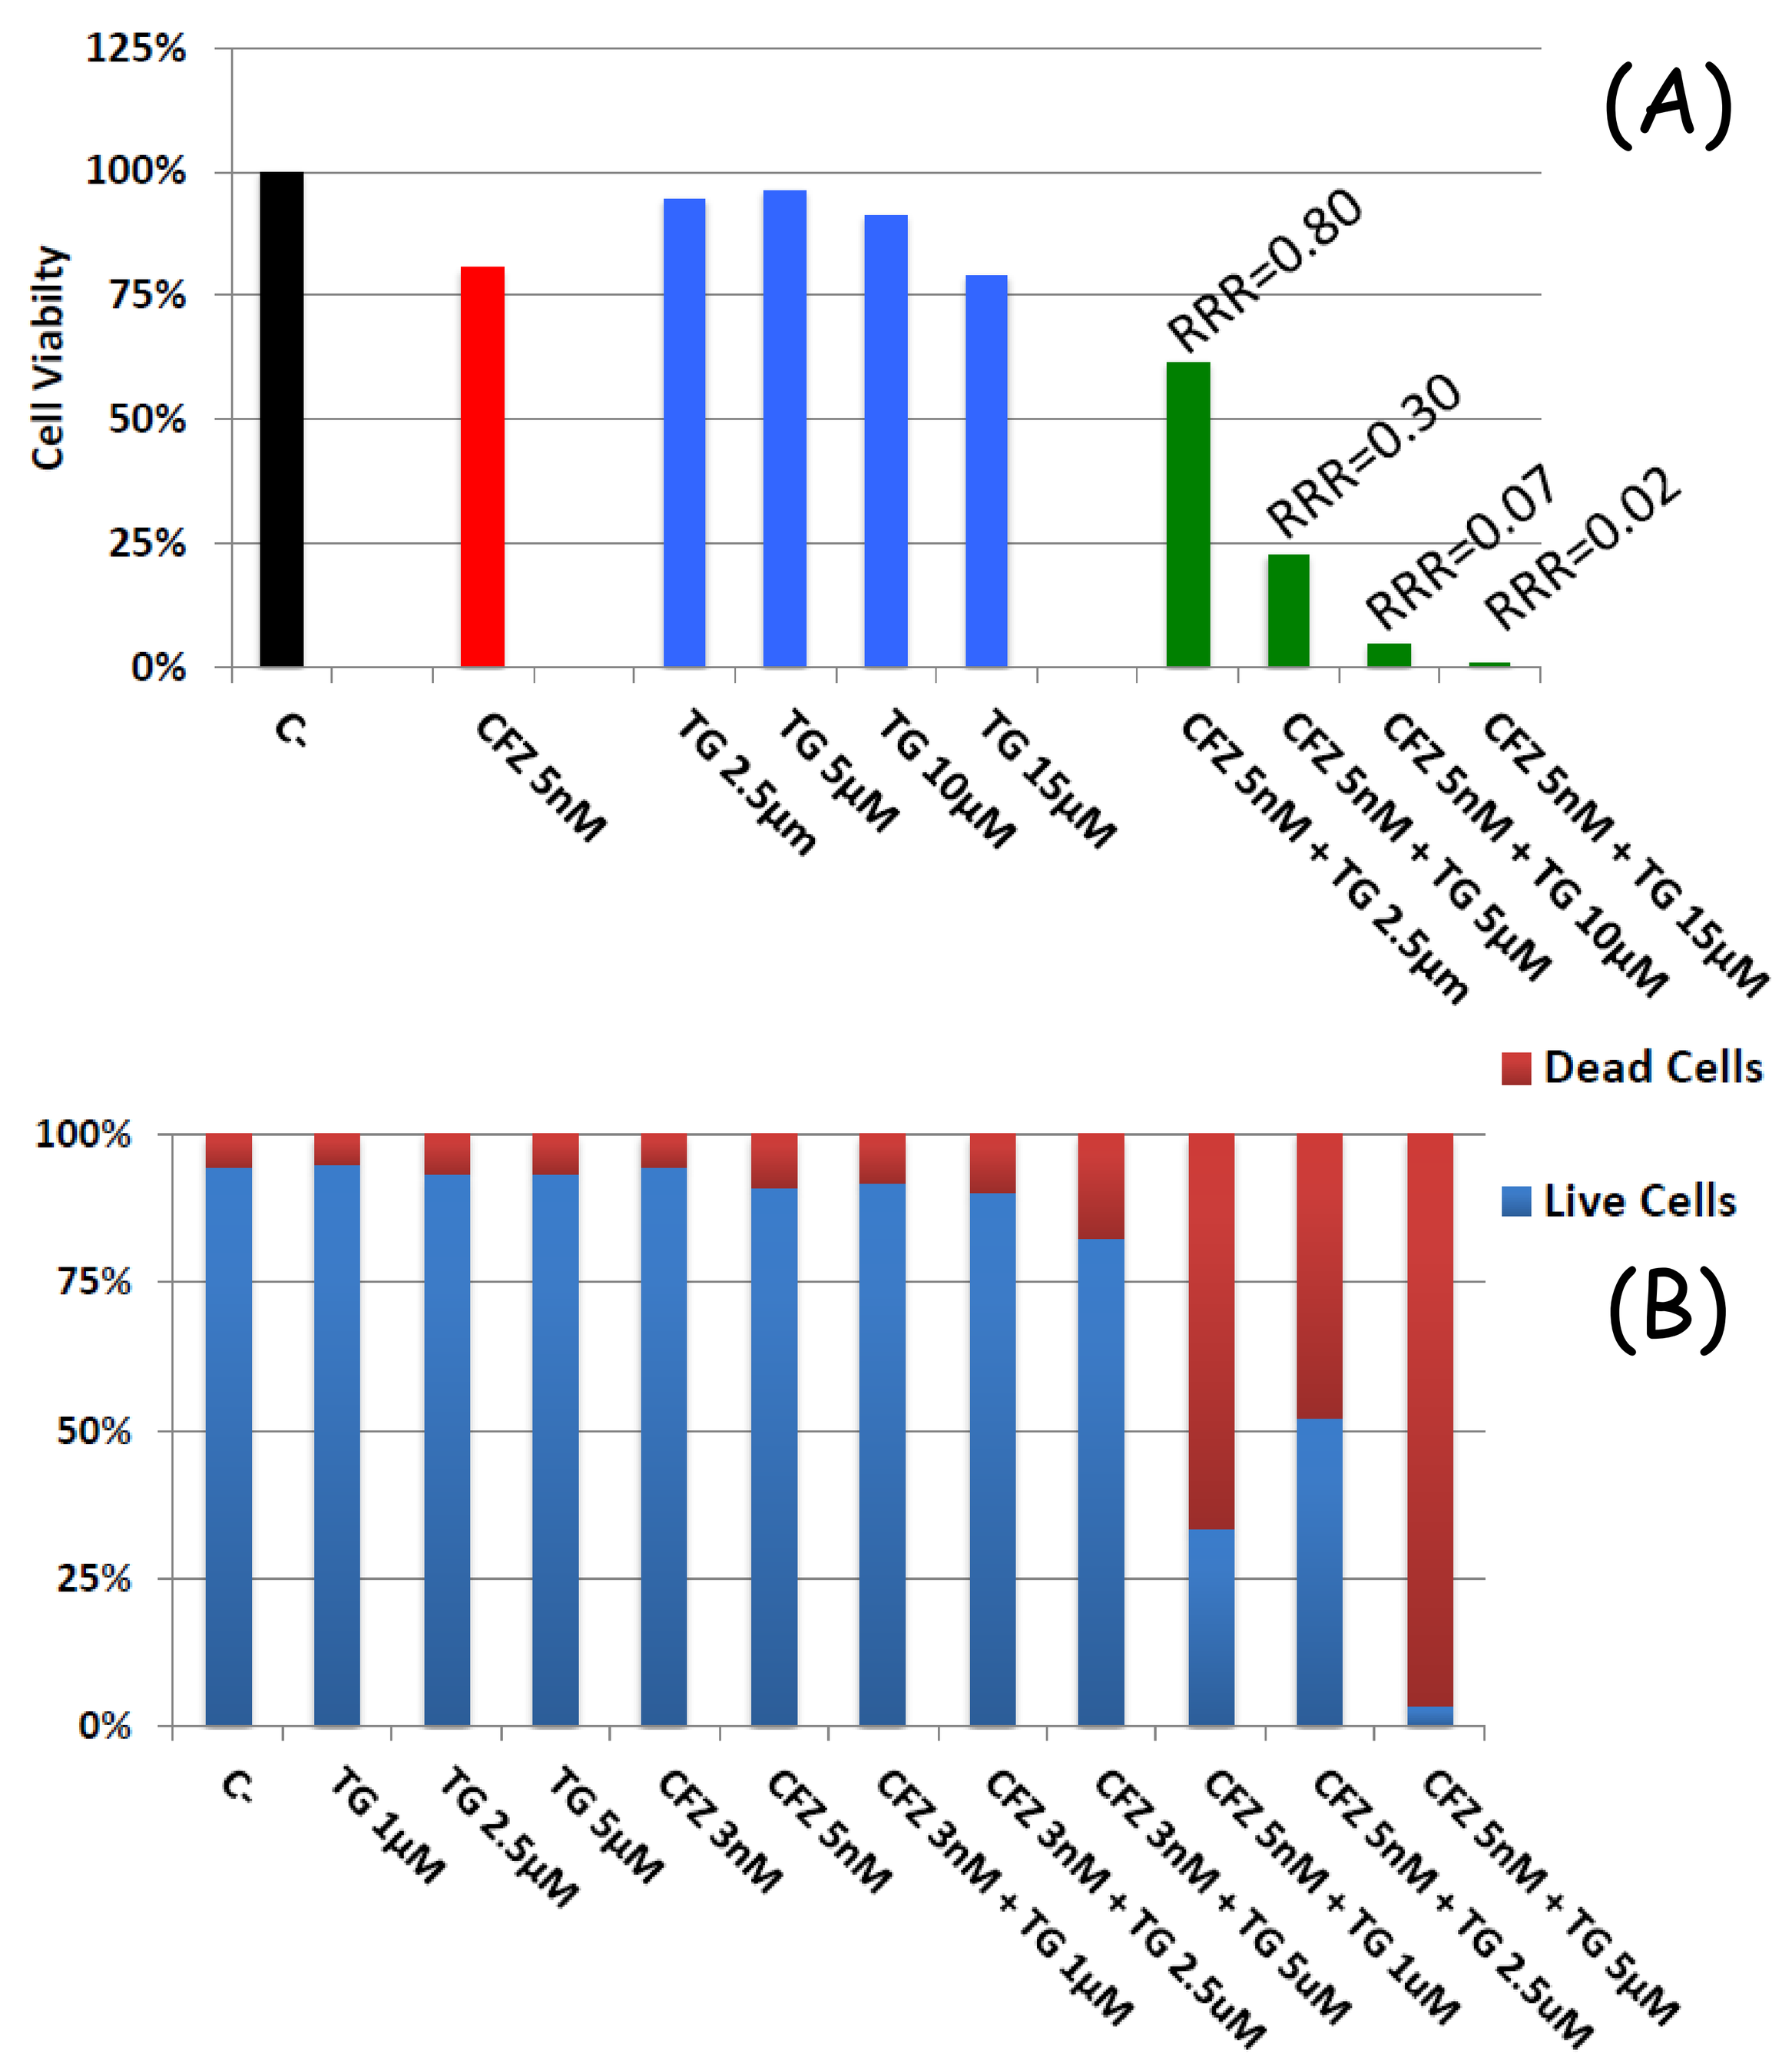

(A)
Cell Viabilty
RRR=0.80
RRR=0.30
RRR=0.07
RRR=0.02
C
CFZ 5nM
TG 2.5µm
TG 5µM
TG 10µM
TG 15µM
CFZ 5nM + TG 2.5µm
CFZ 5nM + TG 5µM
CFZ 5nM + TG 10µM
CFZ 5nM + TG 15µM
(B)
Dead Cells
Live Cells
C
TG 1µM
TG 2.5µM
TG 5µM
CFZ 3nM
CFZ 5nM
CFZ 3nM + TG 1µM
CFZ 3nM + TG 2.5µM
CFZ 3nM + TG 5µM
CFZ 5nM + TG 1uM
CFZ 5nM + TG 2.5uM
CFZ 5nM + TG 5µM

Figure 1. TGR-1202 and carfilzomib were highly synergistic in killing models of cutaneous T cell lymphoma (CTCL).

(A) CTCL cell line H9 was treated with TGR-1202 (TG) or carfilzomib (CFZ) at the indicated concentrations for 48 hours. Cell survival in the treated samples was measured by Cell TiterGlo, and compared with the untreated control. Relative risk ratio (RRR) was calculated to determine synergism. RRR values below 1 indicates synergy.

(B) H9 cells were treated as indicated for 48 hours then prepared for flow cytometry based assay for apoptosis.

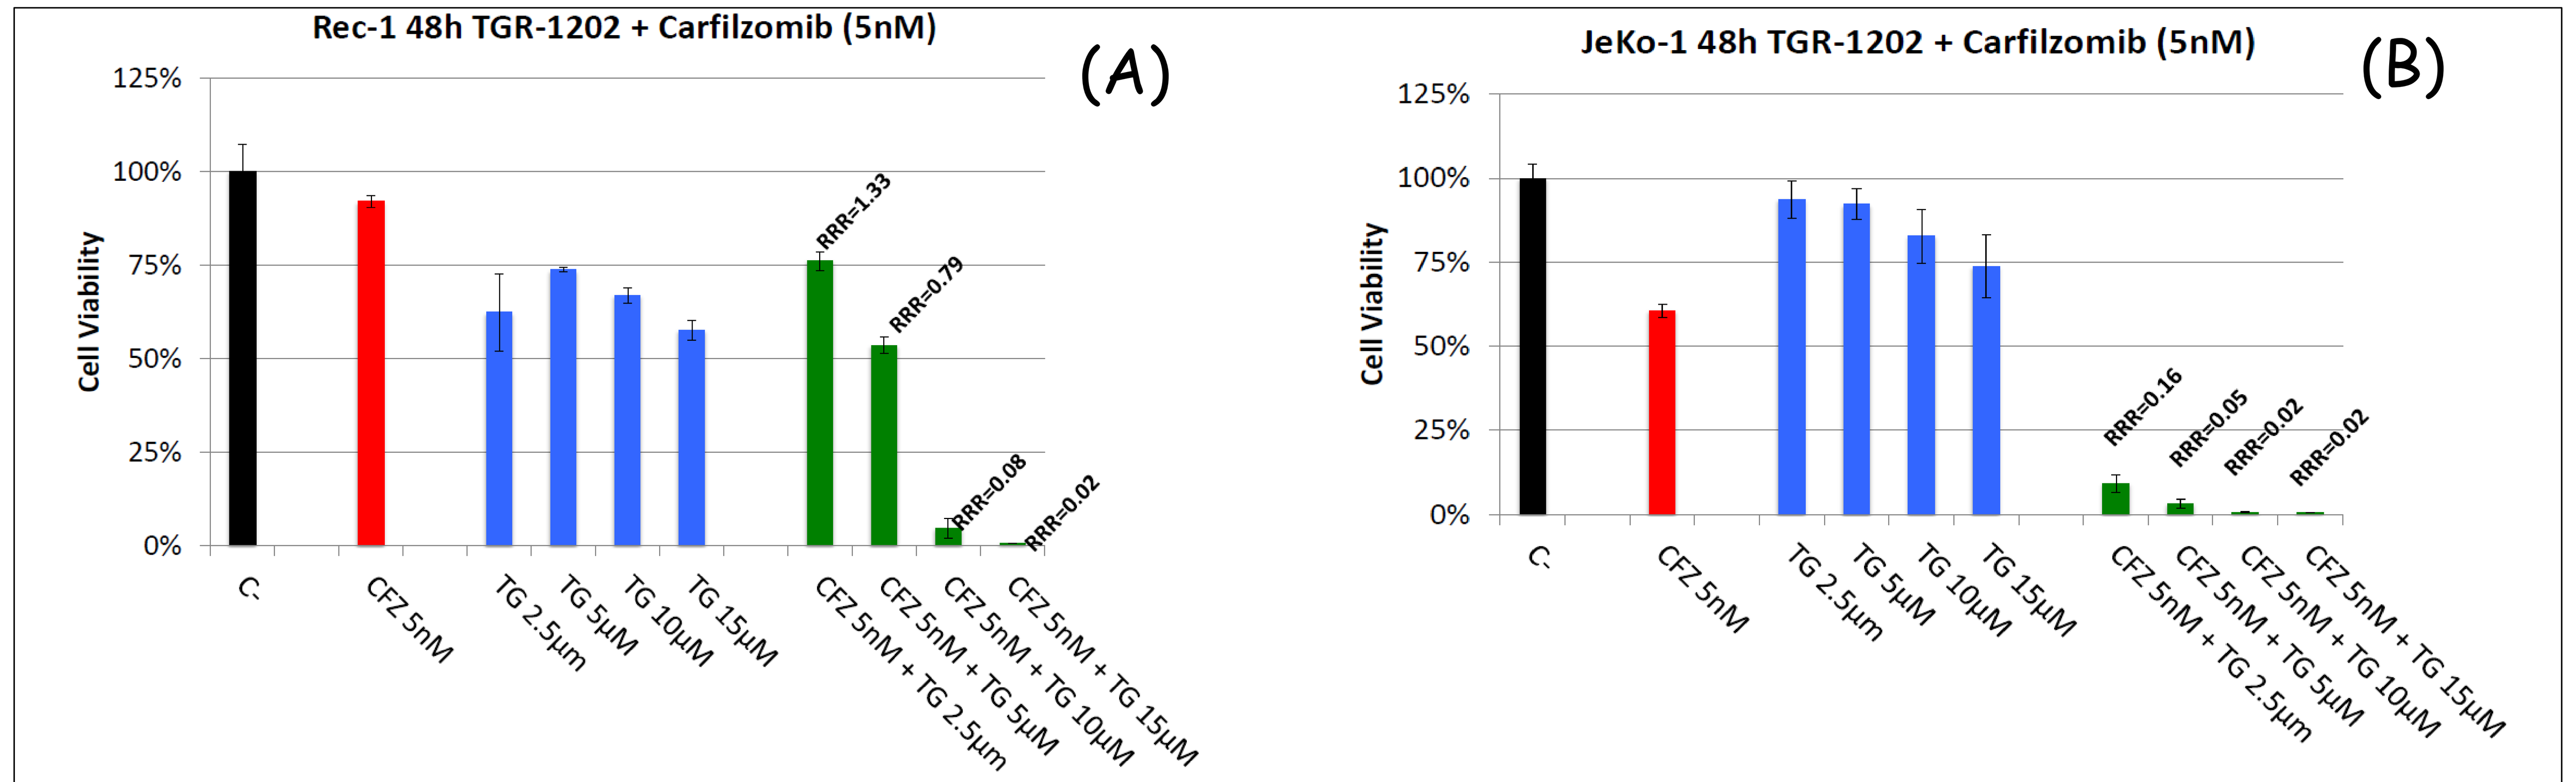

Rec-1 48h TGR-1202 + Carfilzomib (5nM)
(A)
Cell Viability
RRR=1.33
RRR=0.79
RRR=0.08
RRR=0.02
C
CFZ 5nM
TG 2.5µm
TG 5µM
TG 10µM
TG 15µM
CFZ 5nM + TG 2.5µm
CFZ 5nM + TG 5µM
CFZ 5nM + TG 10µM
CFZ 5nM + TG 15µM
JeKo-1 48h TGR-1202 + Carfilzomib (5nM)
(B)
RRR=0.16
RRR=0.05
RRR=0.02
RRR=0.02

Figure 2. TGR-1202 and carfilzomib were highly synergistic in killing models of mantle cell lymphoma (MCL).

MCL cell lines Rec-1 (A) and Jeko-1 (B) were treated with TGR-1202 (TG) or carfilzomib (CFZ) at the indicated concentrations for 48 hours. Cell survival in the treated samples was measured by Cell TiterGlo, and compared with the untreated control. RRR was calculated to determine synergism with RRR values below 1 indicating synergy.

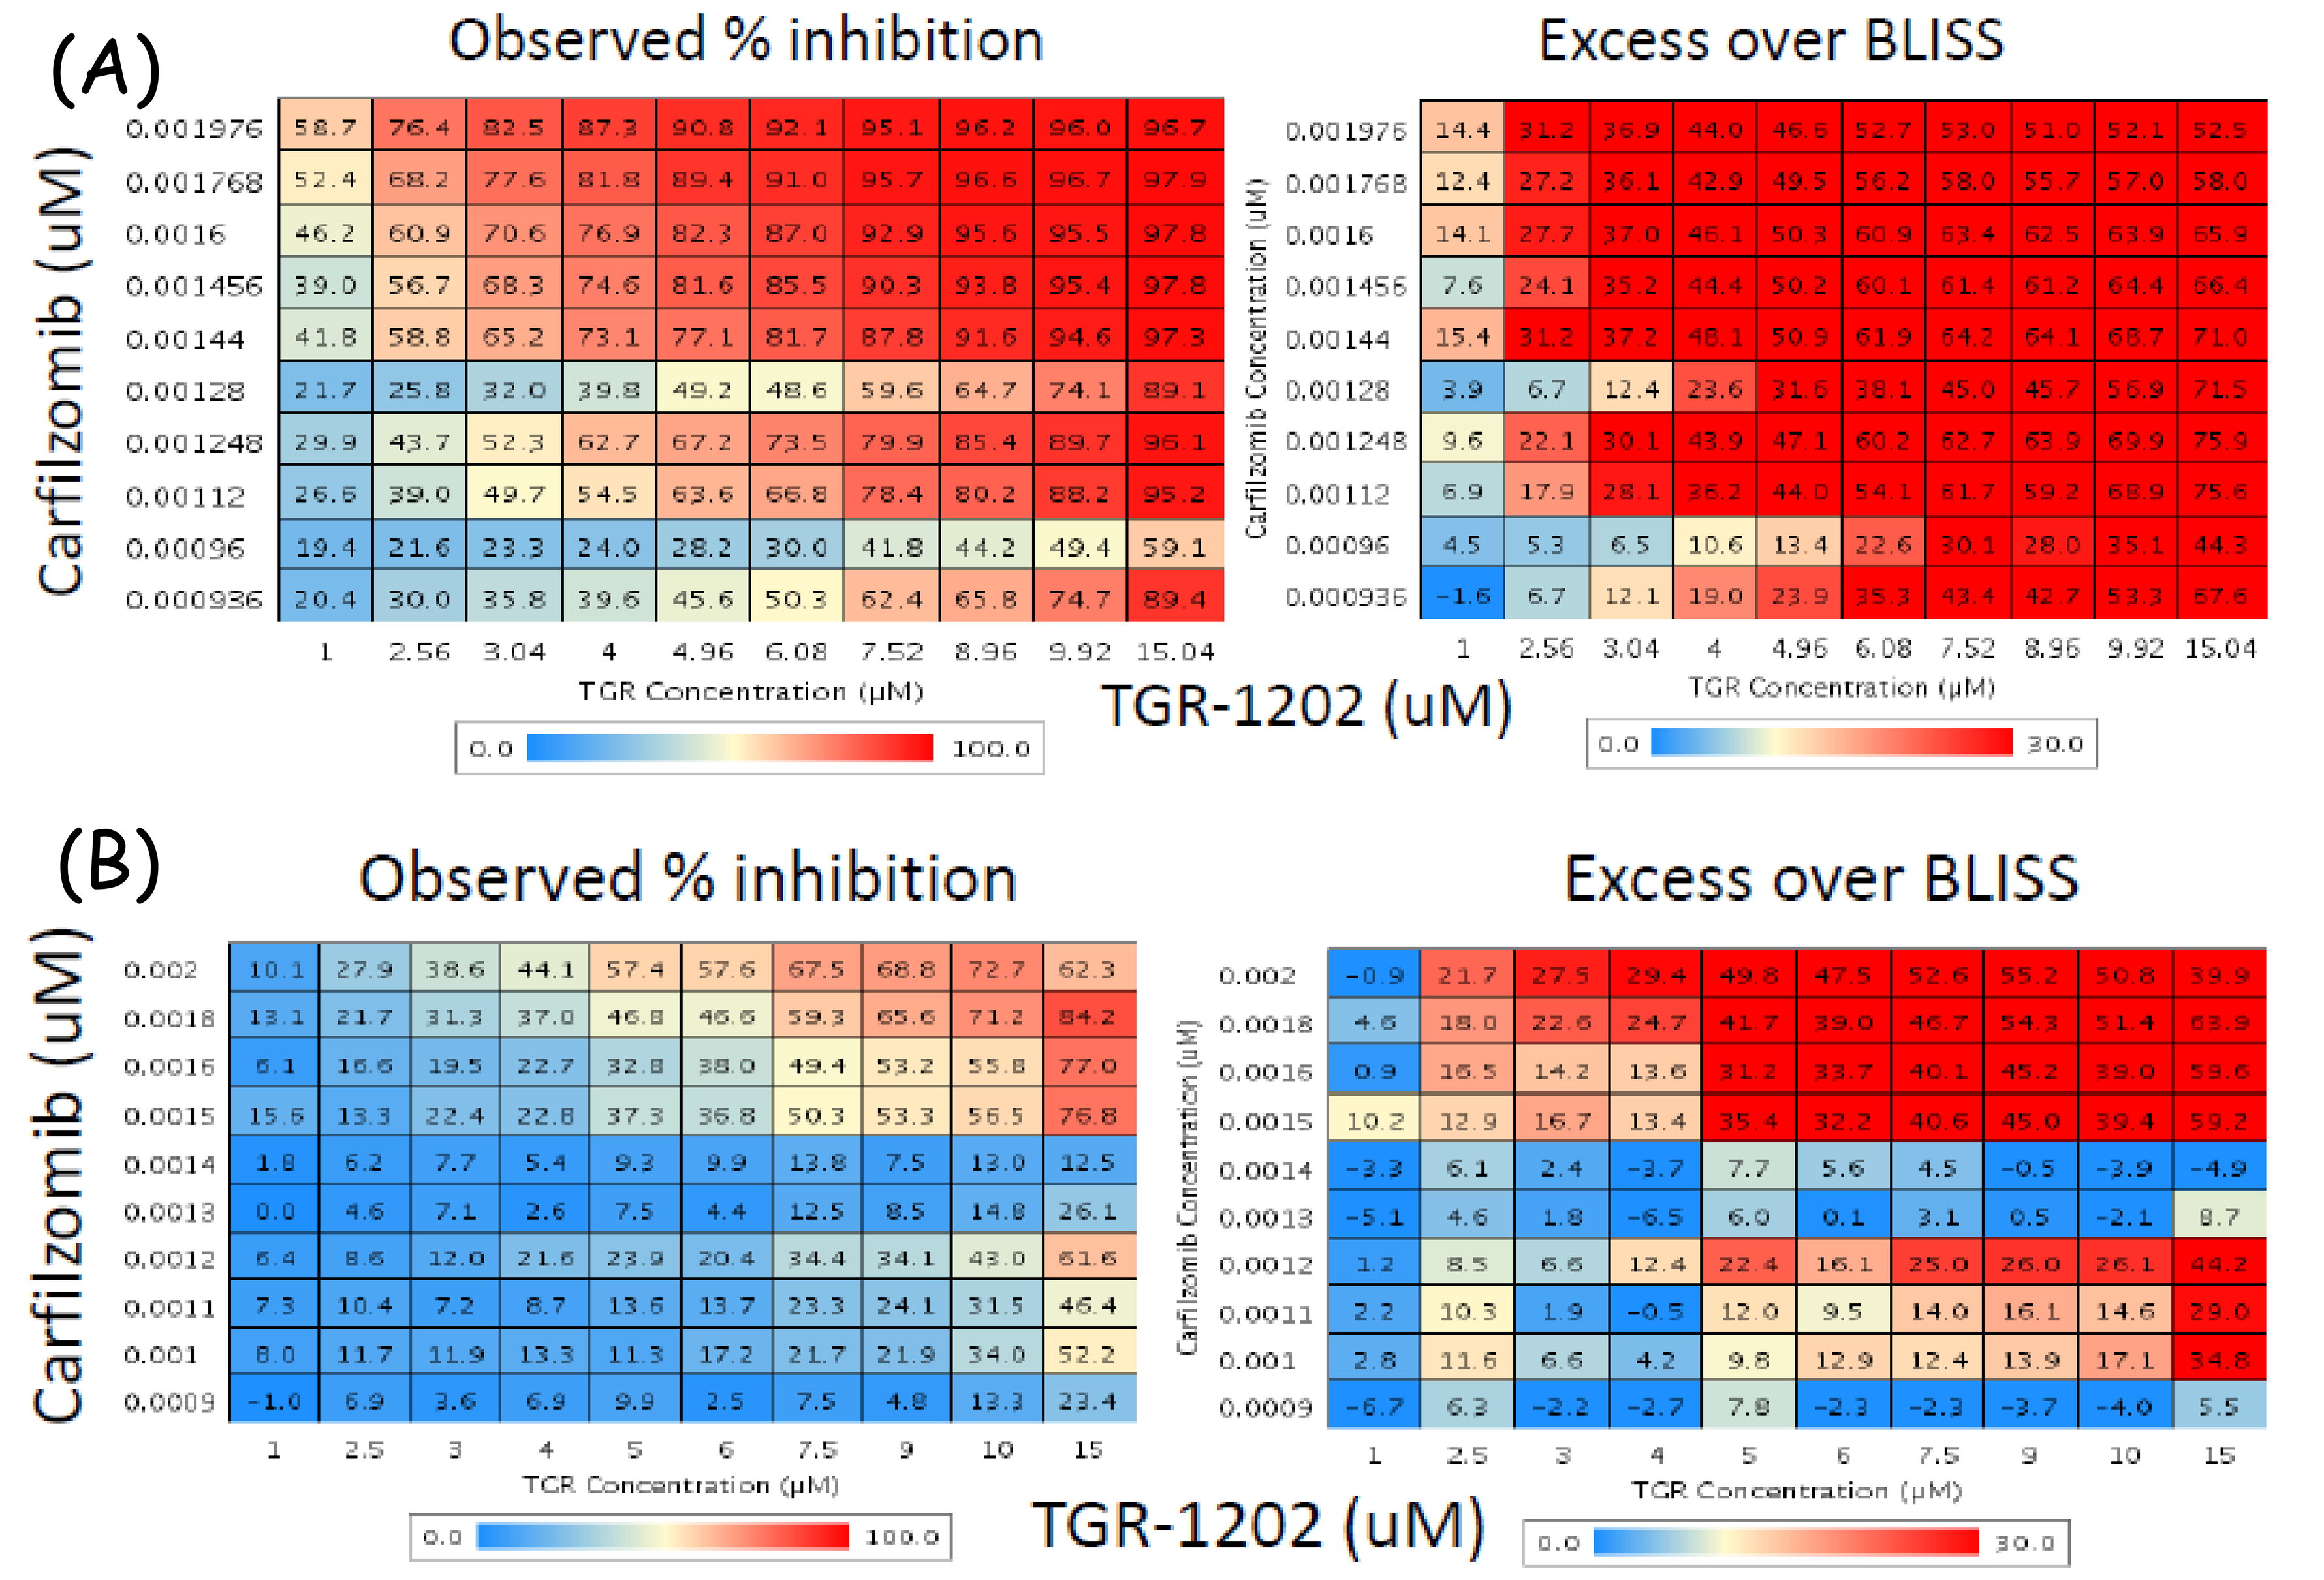

(A)
Observed % inhibition
Excess over BLISS
Carfilzomib (uM)
TGR Concentration (µM)
TGR-1202 (uM)
(B)
Observed % inhibition
Excess over BLISS
Carfilzomib (uM)
TGR Concentration (µM)
TGR-1202 (uM)

Figure 3. TGR-1202 and carfilzomib were highly synergistic in killing models of diffuse large b cell lymphoma (DLBCL).

The DLBCL-ABC cell line OCI-LY10 (A) and GCB cell line SU-DHL4 (B) were treated with TGR-1202 (TG), carfilzomib (CFZ), or the combination for 24 hours using a High Throughput format. Cell survival was measured by Cell TiterGlo. The percentage of inhibition is shown on the left. Excess over BLISS was calculated to determine synergism (Right), with values above 10 indicating synergy.

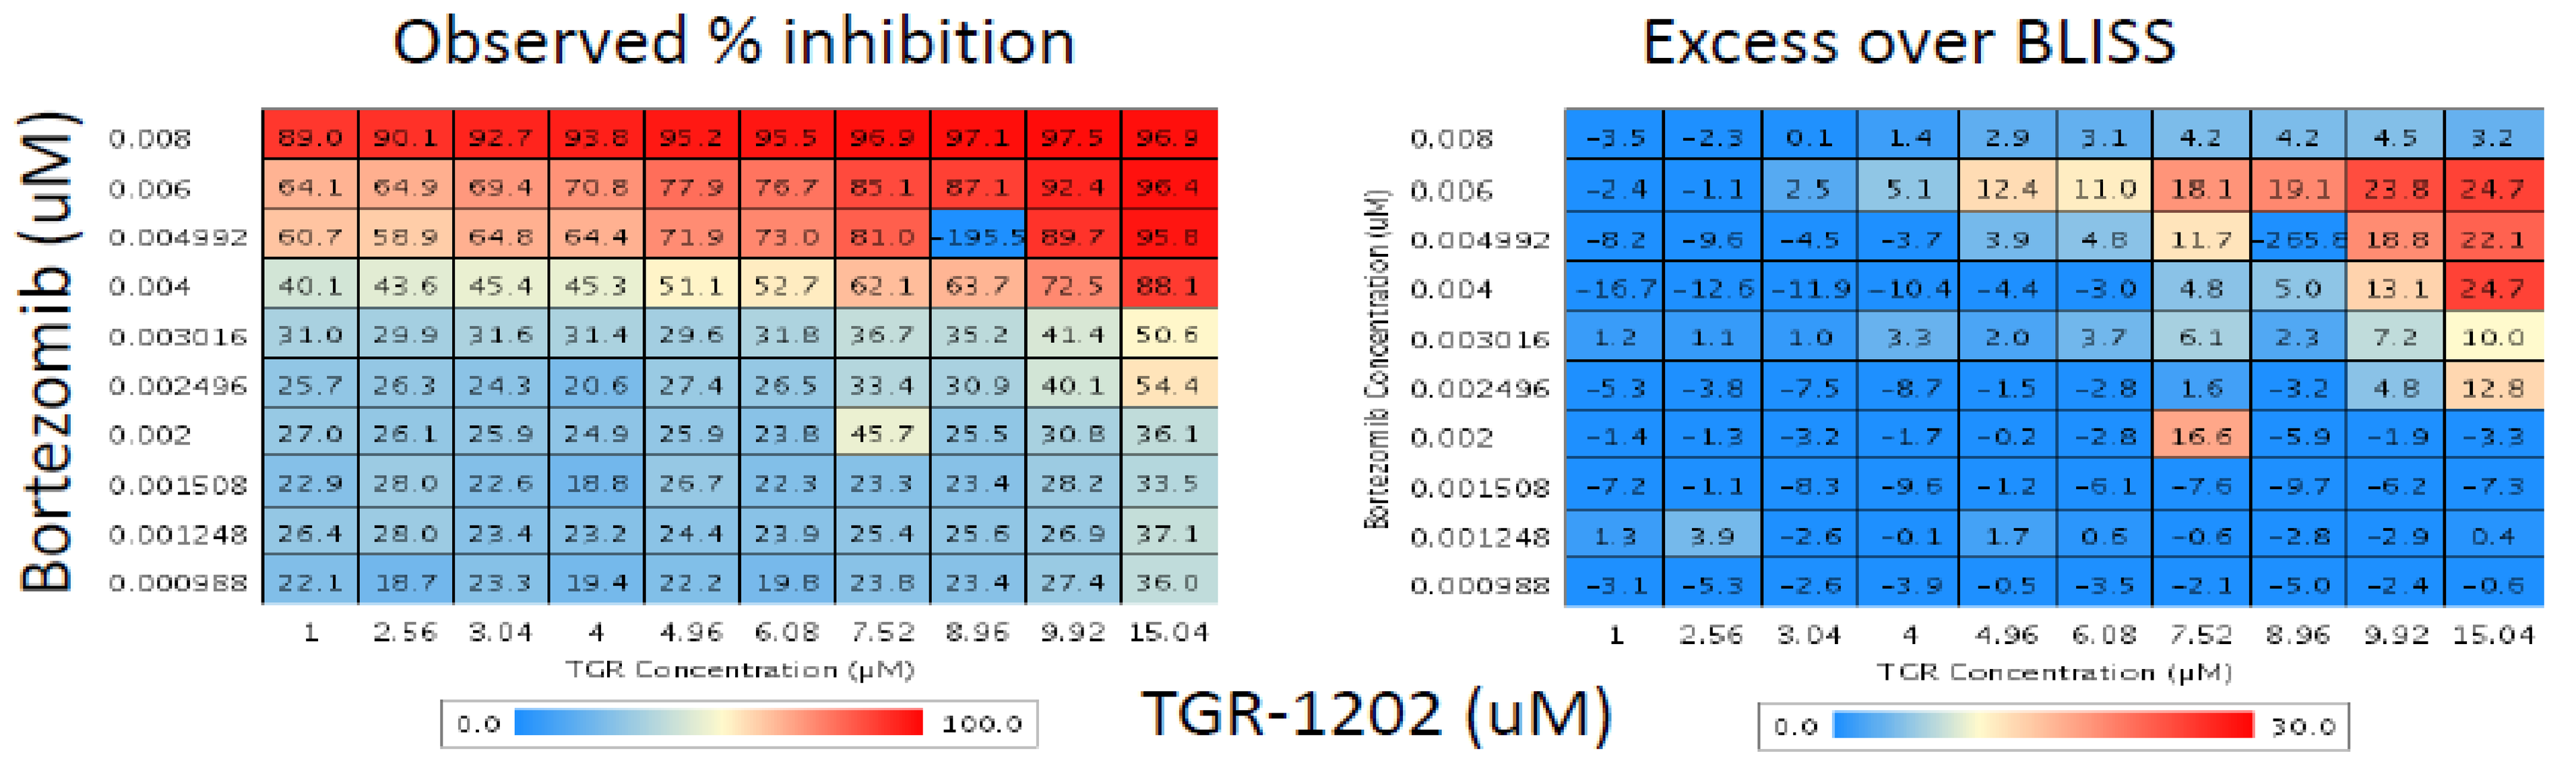

Observed % inhibition
Excess over BLISS
Bortezomib (uM)
TGR Concentration (µM)
TGR-1202 (uM)

Figure 4. TGR-1202 and bortezomib were synergistic but less potent than TGR-1202 and carfilzomib in DLBCL.

DLBCL cell line OCI-LY10 was treated with TGR-1202 (TG), bortezomib, or the combination for 24 hours using a High Throughput format. Inhibition of growth and Excess over BLISS were calculated as in Fig 3.

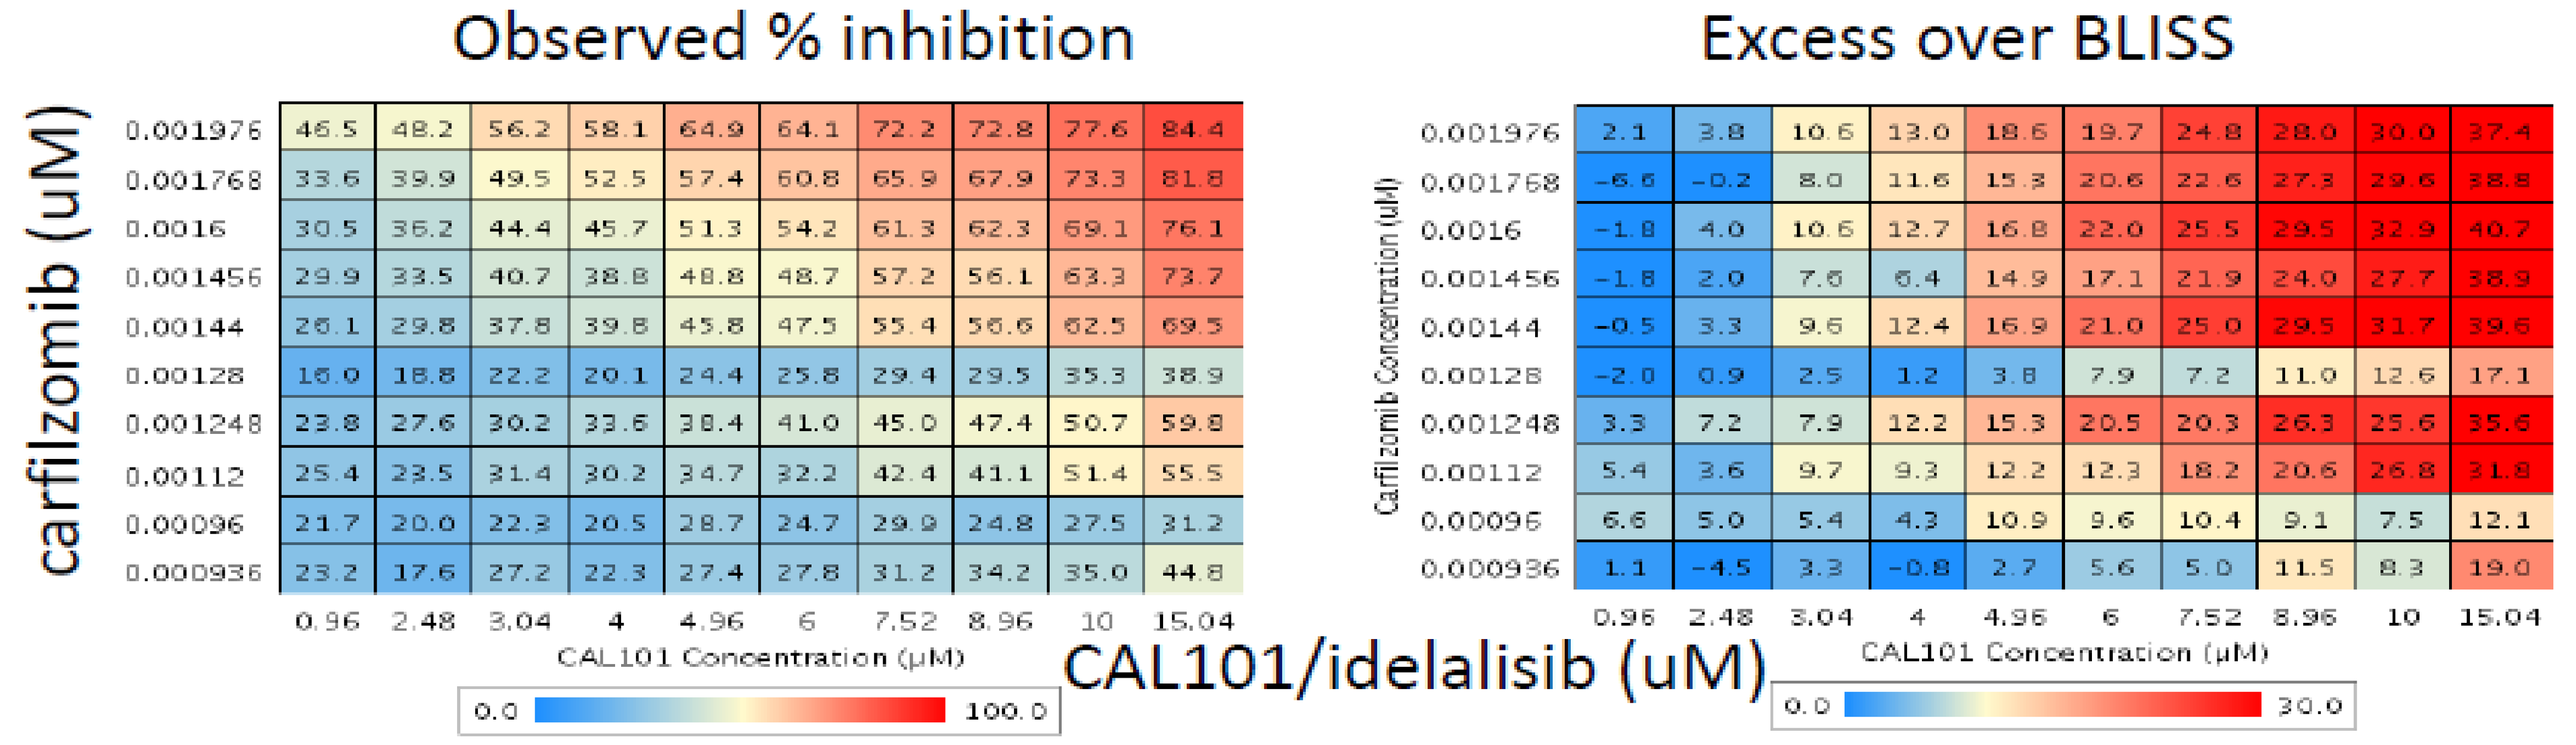

Observed % inhibition
Excess over BLISS
carfilzomib (uM)
CAL101 Concentration (µM)
CAL101/idelalisib (uM)

Figure 5. Carfilzomib and idelalisib were synergistic but less potent than carfilzomib and TGR-1202 in DLBCL.

DLBCL cell line OCI-LY10 was treated with idelalisib, carfilzomib, or the combination for 24 hours using a High Throughput format. Inhibition of growth and Excess over BLISS were calculated as in Fig 3.

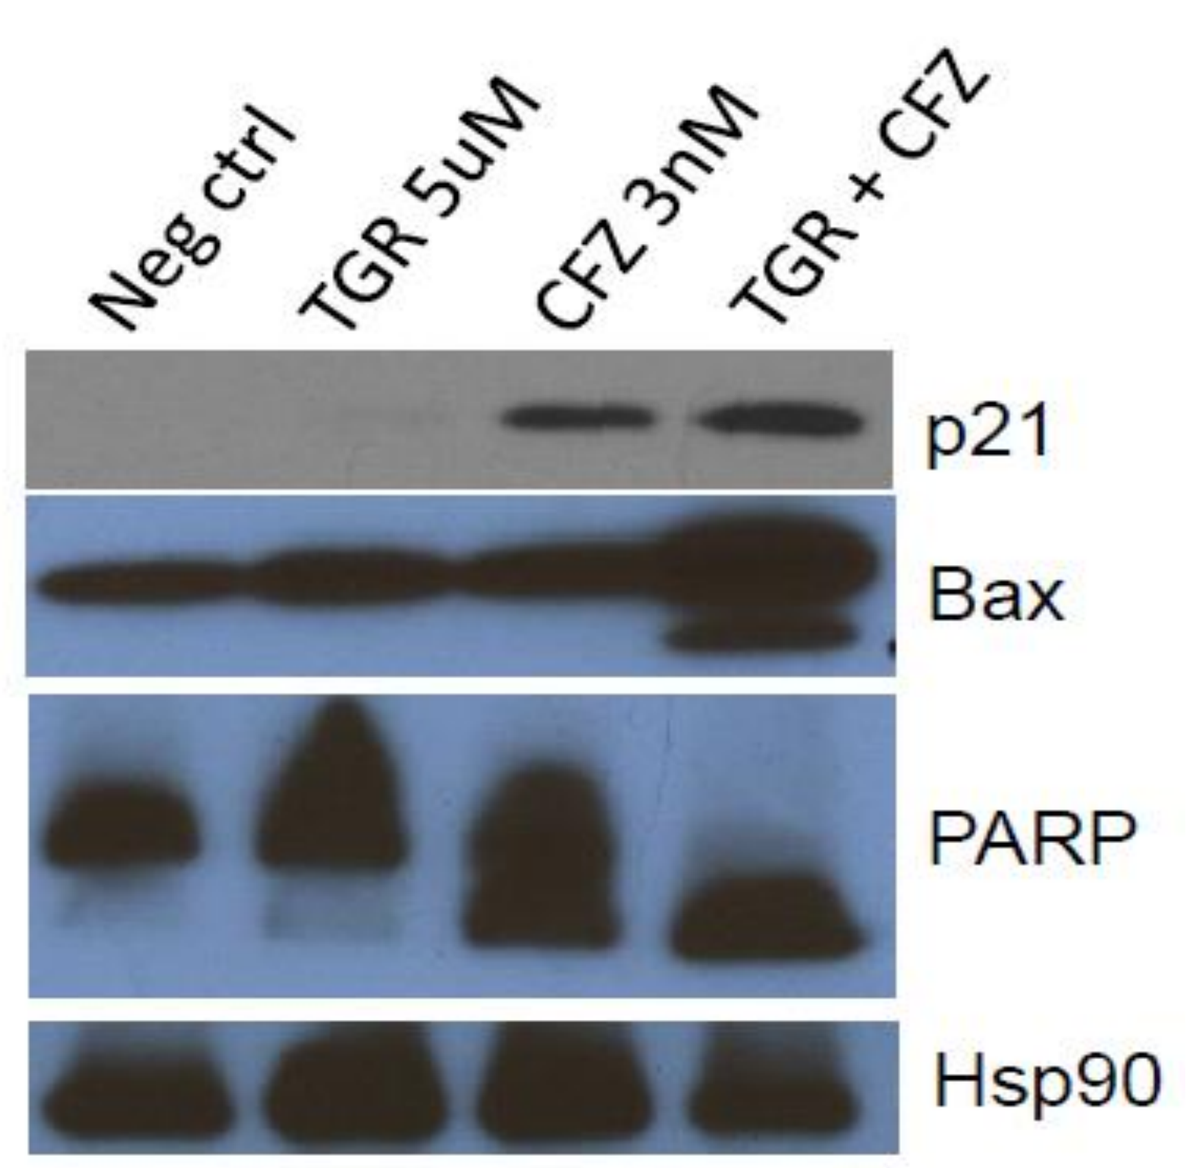

Neg ctrl
TGR 5uM
CFZ 3nM
TGR + CFZ
p21
Bax
PARP
Hsp90

Figure 6. TGR-1202 and carfilzomib synergistically induced the expression of p53 responsive genes and PARP cleavage in models of DLBCL.

DLBCL cell line OCI-LY10 was treated with TGR-1202, carfilzomib, or the combination for 24 hours and processed for Western blot using the indicated antibodies.

CONCLUSIONS

- **TGR-1202 and carfilzomib remarkably and uniquely synergize to kill both B and T-cell lymphoma cells, notably more potently than combinations with other PI3K-δ inhibitors (idelalisib) and/or proteasome inhibitors (bortezomib)**
- **Clinical trials with TGR-1202 in combination with carfilzomib for patients with B and T-cell lymphoma are warranted**

COI: P. Sportelli and H. Miskin (Employment & Equity Ownership – TG Therapeutics, Inc.)
S. Vakkalanka (Employment & Equity Ownership – Rhizen Pharmaceuticals, Inc.)
S. Viswanadha (Employment – Incozen Therapeutics, Inc.)